A First-in-Human Study of RGT-075, a Novel, Orally Bioavailable, Small-Molecule GLP-1 Receptor Agonist, in Healthy Adult Subjects

Regor Pharmaceuticals Inc.

724-P

Mark A. Pirner, Jing Lin, Feng Liu, Lili Yao, Marjorie E. Zettler, David J. Valacer
Regor Pharmaceuticals, Inc., 245 Main Street, Cambridge, MA, USA

ABSTRACT

Glucagon-like peptide-1 receptor agonists (GLP-1 RAs) are a well-characterized and effective class of glucose-lowering agents for the treatment of patients with type 2 diabetes mellitus (T2DM). To date, all market-authorized GLP-1 RAs are peptides, and only one is administered orally. RGT-075 is a novel, orally bioavailable, small-molecule GLP-1 RA which has shown therapeutic potential in animal models of T2DM. To investigate the safety, tolerability, and pharmacokinetics of single escalating oral doses of RGT-075 (15-280 mg) in healthy adult volunteers, a phase 1, randomized, double-blind, placebo-controlled study was conducted. Seven cohorts of 8 subjects were enrolled and randomized to treatment with RGT-075 or matching placebo (6 active/2 placebo per cohort). Plasma and urine samples were collected over 48 hours post-dose and analyzed for RGT-075 concentrations.

A total of 56 healthy adults (64% male, mean age 42.5 years) participated in the study. Treatment-emergent adverse events (TEAEs) were reported by 28 of 56 (50%) subjects (24 subjects treated with RGT-075 and 4 subjects treated with placebo). There were no serious adverse events, deaths, or TEAEs that led to early withdrawal. The majority of TEAEs were considered mild, and the most common TEAEs related to study drug were nausea, vomiting, and headache. Maximum tolerated dose was not reached. Single oral doses of RGT-075 (15-280 mg) resulted in linear increases in C_{max}, AUC_{last}, and AUC_{inf} with ascending doses through 240 mg. Mean elimination half-life ranged from 6.3-11.7 hours. Renal clearance of RGT-075 was low to negligible.

Overall, single RGT-075 doses between 15-280 mg were safe and well-tolerated in healthy volunteers. Further clinical development of RGT-075 as a once-daily, oral therapy for T2DM is underway.

BACKGROUND

- Type 2 diabetes mellitus (T2DM) accounts for over 90 percent of subjects with diabetes. In the US, the prevalence of T2DM is rising in parallel with the increasing prevalence of obesity and sedentary lifestyles.[1]
- GLP-1RAs are effective treatments for patients with T2DM through augmenting insulin secretion and suppressing glucagon release via the stimulation of glucagon-like peptide-1 receptors (GLP-1Rs). GLP-1RAs have been developed and approved as the injectable medication of choice for most subjects who require glucose lowering treatment added to metformin. However, no small molecular GLP-1RAs are yet approved for use in the US, which could present a desirable alternative treatment of T2DM.[2-3]
- RGT-075 is an orally available, small molecular GLP-1RA, which is effective in increasing insulin secretion and reducing glucose levels *in vivo* after both a single-dose and multiple-doses administration.
- This is the first-in-human (FIH), randomized, double-blind, placebo-controlled dose-escalation, single-dose study to evaluate the safety, tolerability, and pharmacokinetics (PK) of a single oral administration of RGT-075 in healthy adult subjects. Herein, we report the safety and PK data of this study.

References

1. American Diabetes Association. Classification and Diagnosis of Diabetes. *Diabetes Care*, 2017, 40 (Suppl 1): S11-S24.
2. Nauck MA; Quast DR; Wefers J; et al. GLP-1 receptor agonists in the treatment of type 2 diabetes - state-of-the-art. *Mol Metab.* 2021, 46:101102.
3. Romera I; Cebrián-Cuenca A; Álvarez-Guisasola F; et al. A Review of PracticalIssueson the Use of Glucagon-Like Peptide-1 Receptor Agonists for the Management ofType 2 Diabetes. *Diabetes Ther*. 2019, 10: 5–19.

STUDY DESIGN

- The study was to consist of up to 8 cohorts comprising up to 64 subjects with 8 subjects per cohort for RGT-075 planned single doses 15-320 mg. Subjects were randomized to either RGT-075 or placebo at a ratio of 3:1. Dose selection followed the protocol-defined dose-escalation plan up to the prespecified maximum dose to be explored.

Screening Period | Treatment Period | Follow-up

Key Eligibility Criteria:
Inclusion: healthy subjects aged 18-65 years, body weight >50 kg, BMI 18.5-29.9 kg/m², baseline vital signs within normal range for age and gender.
Exclusion: history of diabetes, malignancy, multiple endocrine neoplasia 2, medullary thyroid cancer, GI surgery, clinically significant medical history, unwillingness to use appropriate birth control if childbearing potential, GLP-1RA exposure in last 6 months

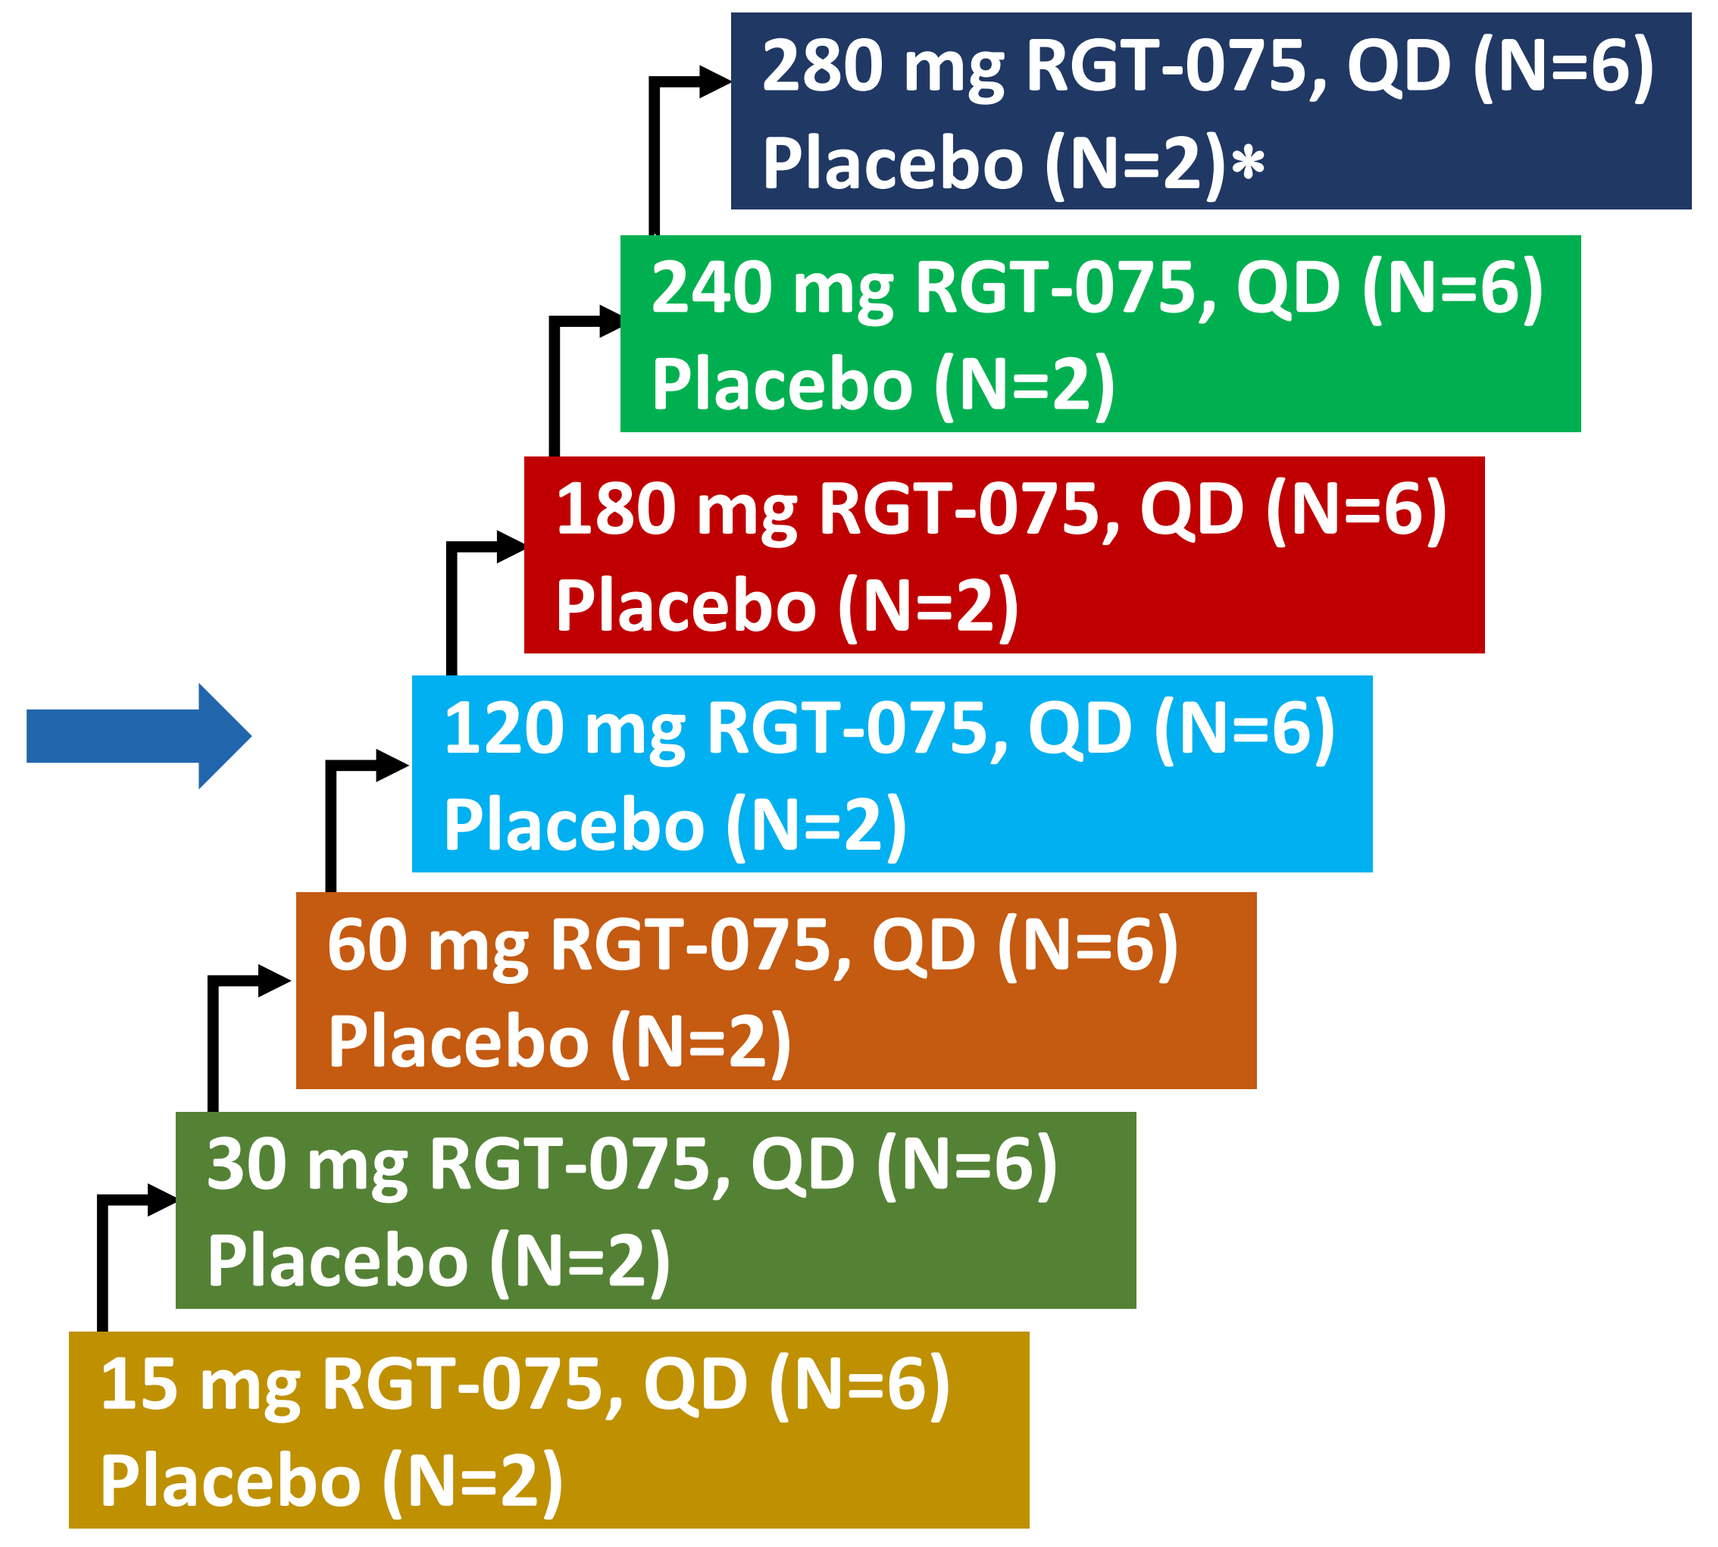
280 mg RGT-075, QD (N=6)
Placebo (N=2)*
240 mg RGT-075, QD (N=6)
Placebo (N=2)
180 mg RGT-075, QD (N=6)
Placebo (N=2)
120 mg RGT-075, QD (N=6)
Placebo (N=2)
60 mg RGT-075, QD (N=6)
Placebo (N=2)
30 mg RGT-075, QD (N=6)
Placebo (N=2)
15 mg RGT-075, QD (N=6)
Placebo (N=2)

Safety follow-up

Primary Objectives:
To assess safety and tolerability of RGT-075 following single-dose administration to healthy adult subjects.

Secondary Objectives:
1. To assess PK of RGT-075 in plasma in healthy adult subjects.
2. To assess urinary excretion of RGT-075 in healthy adult subjects with normal renal function.

BMI, body mass index; GI, gastrointestinal; GLP-1RA, glucagon-like peptide-1 receptor agonist; PK, pharmacokinetics; QD, once daily.
*No subjects were randomized at the dose level of 320 mg since drug exposures (both C_{max} and AUC) didn't appear to increase as the dose increased.

Figure 1 Study Design Schema

RESULTS

Demographics

- In general, the demographic and baseline characteristics were well matched among subjects receiving RGT-075 and between subjects receiving RGT-075 and placebo, with some notable differences among treatment groups related to sex, ethnicity, and race.

Table 1 Demographics and Baseline Characteristics

Characteristics	Cohort 1 RGT-075 15 mg N=6	Cohort 2 RGT- 075 30 mg N=6	Cohort 3 RGT- 075 60 mg N=6	Cohort 4 RGT- 075 120 mg N=6	Cohort 5 RGT- 075 180 mg N=6	Cohort 6 RGT- 075 240 mg N=6	Cohort 7 RGT- 075 280 mg N=6	Pooled Placebo N=14
Age at consent (years)								
Mean (SD)	41.7 (14.7)	48.2 (11.9)	46.2 (12.2)	42.8 (13.6)	42.2 (15.7)	38.5 (11.4)	42.0 (14.8)	40.6 (10.4)
Sex								
Female	2 (33%)	2 (33%)	1 (17%)	4 (67%)	2 (33%)	3 (50%)	3 (50%)	3 (21%)
Ethnicity								
Hispanic or Latino	1 (17%)	2 (33%)	1 (17%)	4 (67%)	2 (33%)	5 (83%)	3 (50%)	5 (36%)
Not Hispanic or Latino	5 (83%)	4 (67%)	5 (83%)	2 (33%)	4 (67%)	1 (17%)	3 (50%)	9 (64%)
Race								
American Indian or Alaska Native	0	1 (17%)	0	0	0	0	0	1 (7%)
Asian	1 (17%)	0	0	0	2 (33%)	0	0	2 (14%)
Black or African American	1 (17%)	4 (67%)	2 (33%)	0	0	1 (17%)	3 (50%)	3 (21%)
White	4 (67%)	1 (17%)	4 (67%)	6 (100%)	4 (67%)	5 (83%)	3 (50%)	10 (71%)
Weight (kg)								
Mean (SD)	73.0 (12.2)	80.7 (7.2)	84.7 (9.0)	71.9 (9.5)	72.3 (10.2)	70.9 (11.5)	73.3 (8.1)	76.5 (11.1)
Height (cm)								
Mean (SD)	169.27 (7.85)	172.0 (11.8)	171.2 (9.7)	168.0 (11.4)	169.2 (10.4)	168.3 (8.7)	171.8 (9.0)	174.8 (8.8)
BMI (kg/m²)								
Mean (SD)	25.3 (2.5)	27.4 (2.9)	28.8 (0.7)	25.5 (2.2)	25.3 (2.6)	25.0 (3.1)	24.9 (2.7)	24.9 (2.2)

SD, standard deviation.

Safety

- No serious adverse events (SAEs), deaths, TEAEs $\geq$ Grade 3, or leading to study discontinuation were reported during the study.
- The most frequently reported TEAEs were GI disorders (nausea and vomiting) and nervous system disorders (headache).
- The incidence of any TEAE appeared to increase with the increase in RGT-075 dose and were reported in 100% of subjects administered single RGT-075 doses ≥180 mg.
- No reported TEAEs related to changes in clinical laboratory evaluations, vital signs, or findings on physical examination.

Table 2 Overall Summary of Treatment-Emergent Adverse Events

TEAE Category, Subjects (%)	Cohort 1 RGT-075 15 mg N=6	Cohort 2 RGT-075 30 mg N=6	Cohort 3 RGT-075 60 mg N=6	Cohort 4 RGT-075 120 mg N=6	Cohort 5 RGT-075 180 mg N=6	Cohort 6 RGT-075 240 mg N=6	Cohort 7 RGT-075 280 mg N=6	Pooled Placebo N=14
Any TEAE	0	2 (33%)	1 (17%)	3 (50%)	6 (100%)	6 (100%)	6 (100%)	4 (29%)
Any Grade 1 TEAE[1]	0	2 (33%)	1 (17%)	3 (50%)	6 (100%)	6 (100%)	6 (100%)	4 (29%)
Any Grade 2 TEAE[1]	0	0	0	1 (17%)	1 (17%)	2 (33%)	1 (17%)	0
TEAEs Related to Study Drug	0	2 (33%)	1 (17%)	3 (50%)	6 (100%)	6 (100%)	6 (100%)	3 (21%)
TEAEs Leading to Study Discontinuation	0	0	0	0	0	0	0	0

TEAE, treatment-emergent adverse event.
1. Grade 1 = mild severity, did not interfere with daily activity; Grade 2 = moderate severity, interfered with daily activity.

Pharmacokinetics

- Single oral doses of RGT-075 resulted in mean C_{max} and AUC increasing in an approximately dose-proportional manner up to 240 mg (Cohort 6). Mean C_{max} and AUC in Cohort 7 (280 mg) did not increase compared to Cohort 6.
- Mean elimination half-life ($T_{1/2}$) was 6.3 to 11.7 hours.
- Mean total body apparent clearance was 4.16 to 7.71 L/h, consistent with a low clearance drug.
- No trends of changes were observed for mean apparent clearance (L/h), or apparent volume of distribution (Vd/F) (L).

Table 3 Plasma PK Parameters

Parameter	Cohort 1 RGT-075 15 mg N=6	Cohort 2 RGT-075 30 mg N=6	Cohort 3 RGT-075 60 mg N=6	Cohort 4 RGT-075 120 mg N=6	Cohort 5 RGT-075 180 mg N=6	Cohort 6 RGT-075 240 mg N=6	Cohort 7 RGT-075 280 mg N=6
T_{max} (h)	2.17 (0.41)	3. 33 (2.07)	5.85 (8.90)	2.68 (0.53)	3.17 (1.60)	7.20 (8.59)	3.33 (1.37)
C_{max} (ng/mL)	333.83 (43.53)	615.83 (164.50)	809.33 (312.90)	1947.67 (999.47)	2413.33 (1364.52)	3695.00 (1245.24)	3581.67 (1412.48)
AUC_{last} (ng*h/mL)	2524.81 (664.15)	7395.08 (1549.12)	14150.39 (10800.46)	27272.37 (9053.94)	38191.74 (5991.12)	63581.06 (29700.00)	49206.21 (25764.23)
AUC_{inf} (ng*h/mL)	2546.11 (664.10)	7510.46 (1586.54)	10056.62 (3801.49)[a]	28286.23 (9831.58)	36717.86 (5557.56)[b]	68632.17 (39987.20)[b]	42968.02 (22025.25)[a]
Half-life ($t_{1/2}$) (h)	6.26 (1.02)	6.90 (1.55)	6.50 (0.97)[a]	8.33 (2.99)	11.73 (7.35)[a]	7.68 (2.44)[b]	8.19 (3.65)[a]
Apparent Clearance (L/h)	6.20 (1.43)	4.16 (0.95)	6.67 (2.38)[a]	4.92 (2.39)	5.00 (0.87)[b]	5.36 (4.65)[b]	7.71 (3.02)[a]
Apparent Volume of Distribution (L)	55.46 (14.40)	39.83 (5.71)	60.48 (15.38)[a]	53.67 (14.77)	60.72 (8.85)[b]	50.03 (30.36)[b]	80.72 (16.34)[a]

AUC, area under the curve; CV, coefficient of variation; max, maximum; SD, standard deviation.
Data are shown as Mean (SD), [a]n=5, [b]n=4.

- RGT-075 PK analyses demonstrated dose proportionality through the 240 mg dose.

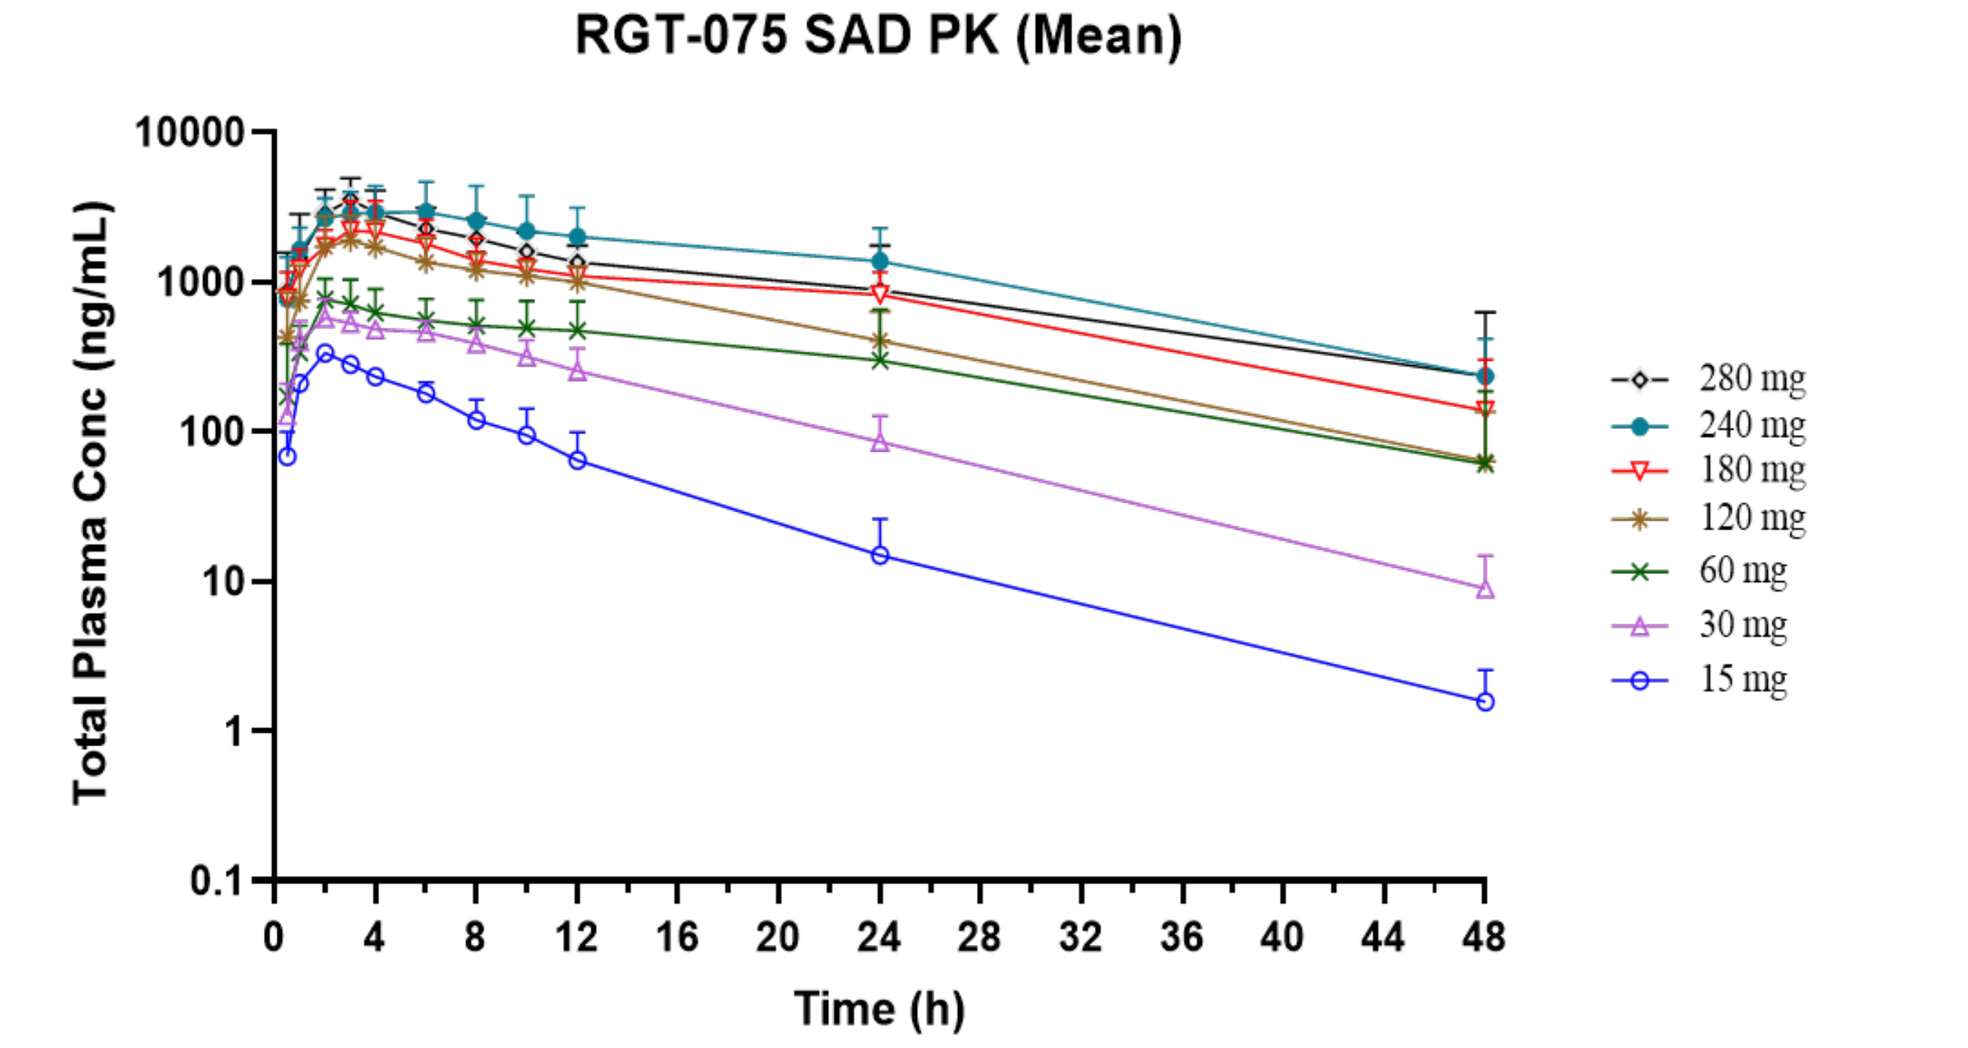
RGT-075 SAD PK (Mean)
Total Plasma Conc (ng/mL)
10000
1000
100
10
1
0.1
0
4
8
12
16
20
24
28
32
36
40
44
48
Time (h)
280 mg
240 mg
180 mg
120 mg
60 mg
30 mg
15 mg

Figure 2 Mean (+SD) RGT-075 Plasma Concentration Time Profiles (Semi-log Scale) following Single Oral Dose of RGT-075 to Healthy Volunteers

CONCLUSIONS

- Overall, single ascending oral doses of RGT-075 from 15 to 280 mg were safe in healthy adult subjects.
- RGT-075 exposures were dose-proportional up to 240 mg but did not increase above 240 mg levels.
- RGT-075 PK demonstrated low total body clearance and a reasonably good distribution throughout the body.
- The mean elimination half-life ($t_{1/2}$) (from 6.3 to 11.7 hours) indicated suitability for QD dosing.
- Renal clearance was very low to negligible (<0.01% of the total dosed RGT-075).
- These results support advancing RGT-075 clinical development to the evaluation of multiple ascending doses.